A Multicenter, Phase 2, Dose-Escalation/Dose-Expansion Study of Dabogratinib (Formerly TYRA-300) in Children With Achondroplasia With Open Growth Plates: BEACH301

SAT-474

Klane White,[1] Ricki S. Carroll,[2] Michael Stalvey,[3] Carl Decker,[3] Andrew Barrett,[3] Will Charlton,[4] Ravi Savarirayan[5]

[1]Children's Hospital Colorado, Denver, CO; [2]Nemours Children's Hospital, Wilmington, DE; [3]Tyra Biosciences, Inc., Carlsbad, CA; [4]Work completed while the author was an employee of Tyra Biosciences, Inc., Carlsbad, CA, USA; [5]Murdoch Children's Research Institute, Royal Children's Hospital, Melbourne, VIC, Australia.

BACKGROUND

- Achondroplasia is the most common form of heritable skeletal dysplasia, affecting approximately 250,000 people worldwide[1,2]
- Achondroplasia is caused by a gain of function mutation in the *FGFR3* gene (most commonly G380R) leading to overactivation of FGFR3 signaling[1-4]
- Aberrant FGFR3 signaling inhibits chondrocyte proliferation and differentiation, resulting in impaired endochondral ossification and abnormal skeletal development (Figure 1)[2,3,5]
- Individuals with achondroplasia can experience lifelong morbidity, with early-life complications including foramen magnum stenosis, recurrent otitis media, sleep-disordered breathing, and ongoing impact across childhood and adulthood including obesity, skeletal complications, chronic pain, surgeries, and effects on function and psychosocial well-being[1,3,6,7]
- Inhibiting FGFR3 may benefit children with achondroplasia. Approved CNP analogs indirectly modulate this pathway,[2,8,9] while pan-FGFR inhibition has demonstrated encouraging safety and efficacy, supporting direct FGFR inhibition[10]
 - Dabogratinib is an oral, highly selective FGFR3 inhibitor designed to directly target the underlying driver of achondroplasia while minimizing off-target effects[11]
 - By avoiding the toxicities associated with inhibition of FGFR1, FGFR2, and FGFR4, dabogratinib may provide greater efficacy with a larger therapeutic window than non-isoform selective FGFR inhibitors[12]
 - Dabogratinib is being evaluated in children with achondroplasia in the phase 2 BEACH301 study (NCT06842355)[13]

Figure 1. Aberrant FGFR3 Signaling in Achondroplasia

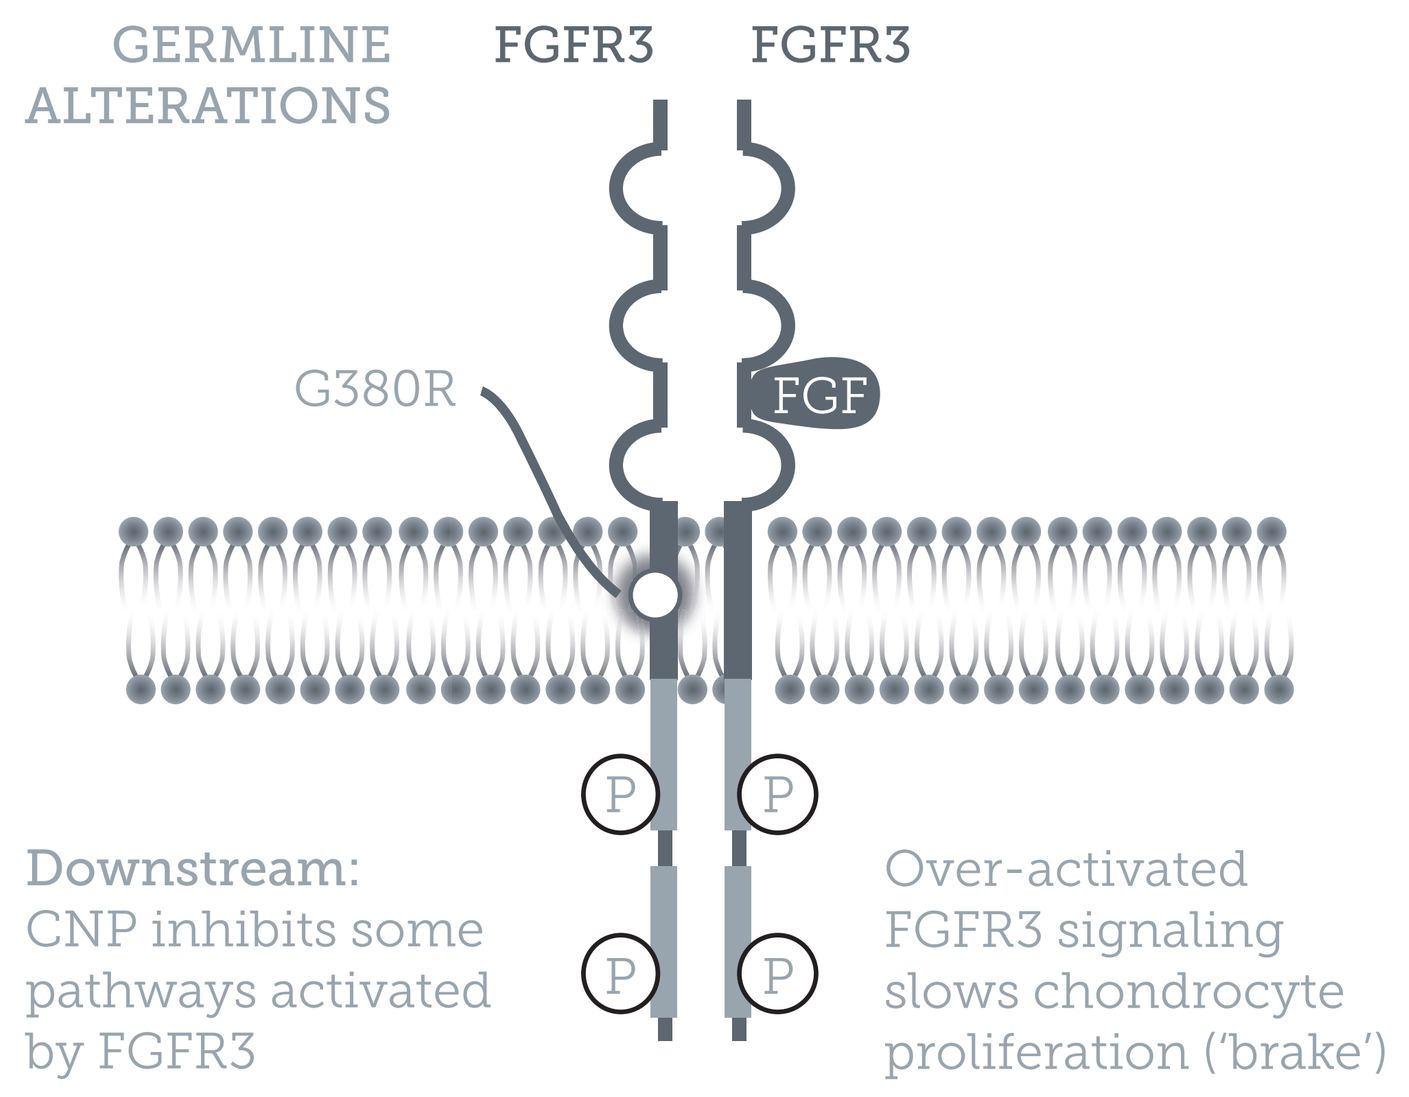

GERMLINE ALTERATIONS
FGFR3
FGFR3
G380R
FGF
P
Downstream:
CNP inhibits some pathways activated by FGFR3
Over-activated FGFR3 signaling slows chondrocyte proliferation ('brake')

Gain-of-function *FGFR3* mutations (eg, G380R) lead to overactivation of FGFR3 signaling (via an increase in steady-state levels of receptor),[4] inhibiting chondrocyte proliferation and impairing endochondral bone growth.

METHODS

- BEACH301 (NCT06842355) is a phase 2, multicenter, open-label, dose-escalation/dose-expansion study to evaluate the safety, tolerability, and potentially effective dose(s) of oral dabogratinib QD in children aged 3 to 10 years with achondroplasia with open growth plates[13]
- Dabogratinib is administered at weight-based dose levels (0.125-0.625 mg/kg), with dose escalation per protocol-defined criteria
- The study includes 3 cohorts (Figure 2)
 - Sentinel Safety Cohort
 - Cohort 1: Treatment-naïve
 - Cohort 2: Received prior growth-accelerating therapy

Figure 2. Study Design of BEACH301

Sentinel Safety Cohort
ACH age 5-10
No run-in
Anticipate n=~25 total enrollment

	DOSE (mg/kg)
✓	0.625
✓	0.50
✓	0.375
✓	0.25
✓	0.125

Cohorts 1 and 2*

N=*	DOSE (mg/kg)
6	0.625
6	0.50
6	0.375
6	0.25
6	0.125

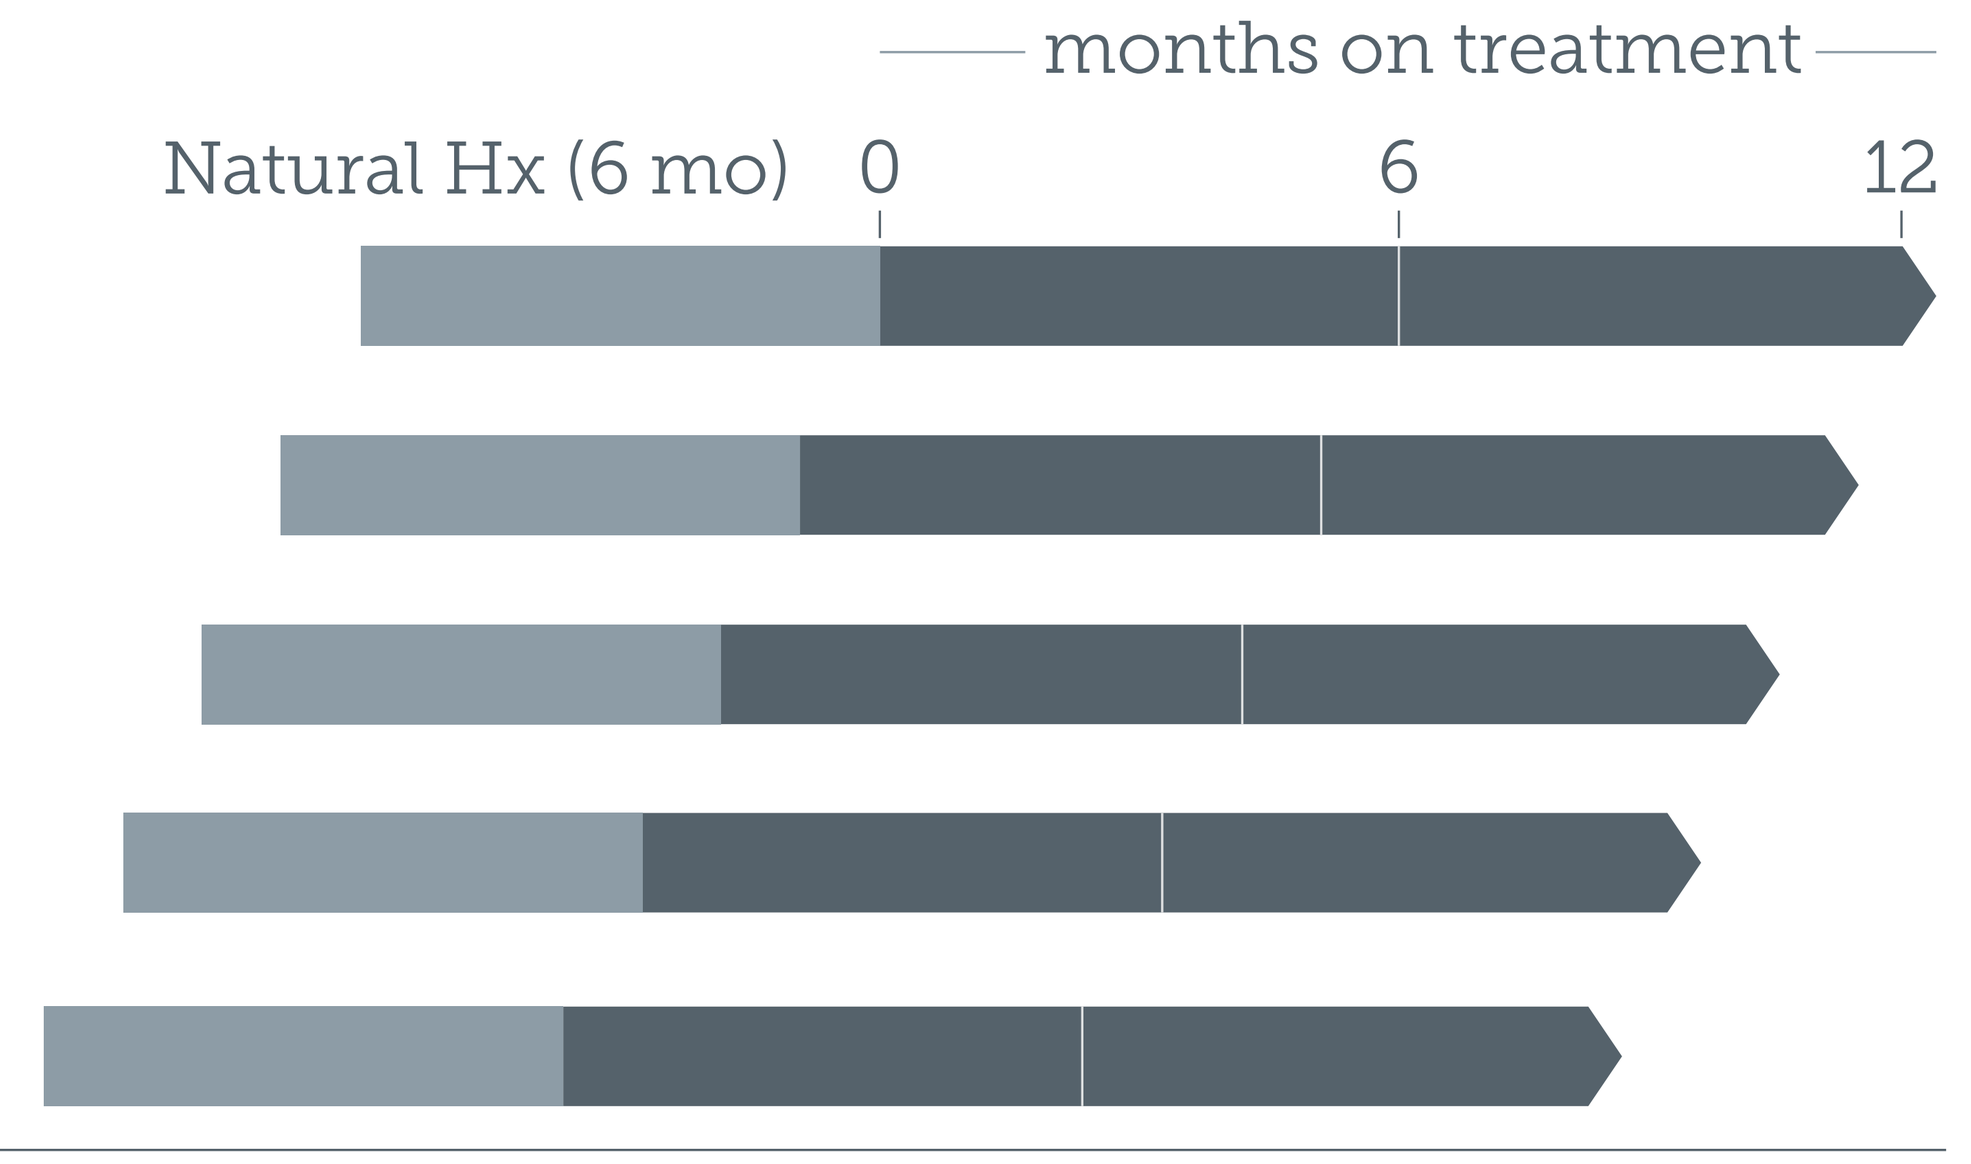

months on treatment
Natural Hx (6 mo)
0
6
12

*Cohort 1 (6-mo run-in)
Treatment naïve;
ACH age 3-10

*Cohort 2 (6-mo run-in)
Received prior growth-accelerating therapy; ACH age 3-10

✓ Indicates that the dose level has been cleared by an independent Data Safety Monitoring Committee.*Dose decisions based on 6 participants. Additional participants may be assigned at discretion of Sponsor.
Schematic representation of the study design, illustrating cohort structure and weight-based dabogratinib dosing.

Participant Criteria[13]

Key Inclusion Criteria	Key Exclusion Criteria
• Children aged 3 to 10 years of age • Molecularly confirmed achondroplasia with *FGFR3* G380R mutation • Radiographically confirmed open growth plates by bone age X-ray • Ability to stand and ambulate independently • Sentinel Safety Cohort only: aged 5 to 10 years old (inclusive) • Cohort 1 only: aged 3 to 10 years old (inclusive) and are naïve to prior growth-accelerating therapy • Cohort 2 only: aged 3 to 10 years old (inclusive) and have received prior growth-accelerating therapy • Ability to take oral medication	• Presence or history of any concurrent disease or condition that would interfere with study participation, safety evaluations, or any uncontrolled or untreated condition that could impact pediatric growth • Endocrine condition that alters calcium/phosphate homeostasis • Prior limb lengthening surgery or planned or expected to have limb lengthening surgery while enrolled in the study • History or current evidence of corneal or retinal disorder/keratopathy • Presence of guided growth hardware (eg, 8 plates). Planned or anticipated orthopedic surgeries

Study Endpoints[13]

Primary Endpoints	Secondary Endpoints
• Incidence of treatment-emergent AEs • Change from baseline in AGV	• Change from baseline in height z-score • Comparison of single- and multiple-dose sparse pharmacokinetic parameters, including but not limited to C_{max}, T_{max}, $AUC_{0\text{-last}}$, AUC_{Tau}, $AUC_{0-\infty}$, Vd/F, CL/F, and $t_{½}$ • Change from baseline in anthropometric measurements, including but not limited to standing height, sitting height, upper arm length, lower arm length, tibial length, femur length, arm span proportionality (arm span/height ratio), and upper segment/lower segment ratio with dabogratinib

BEACH301 SITE LIST

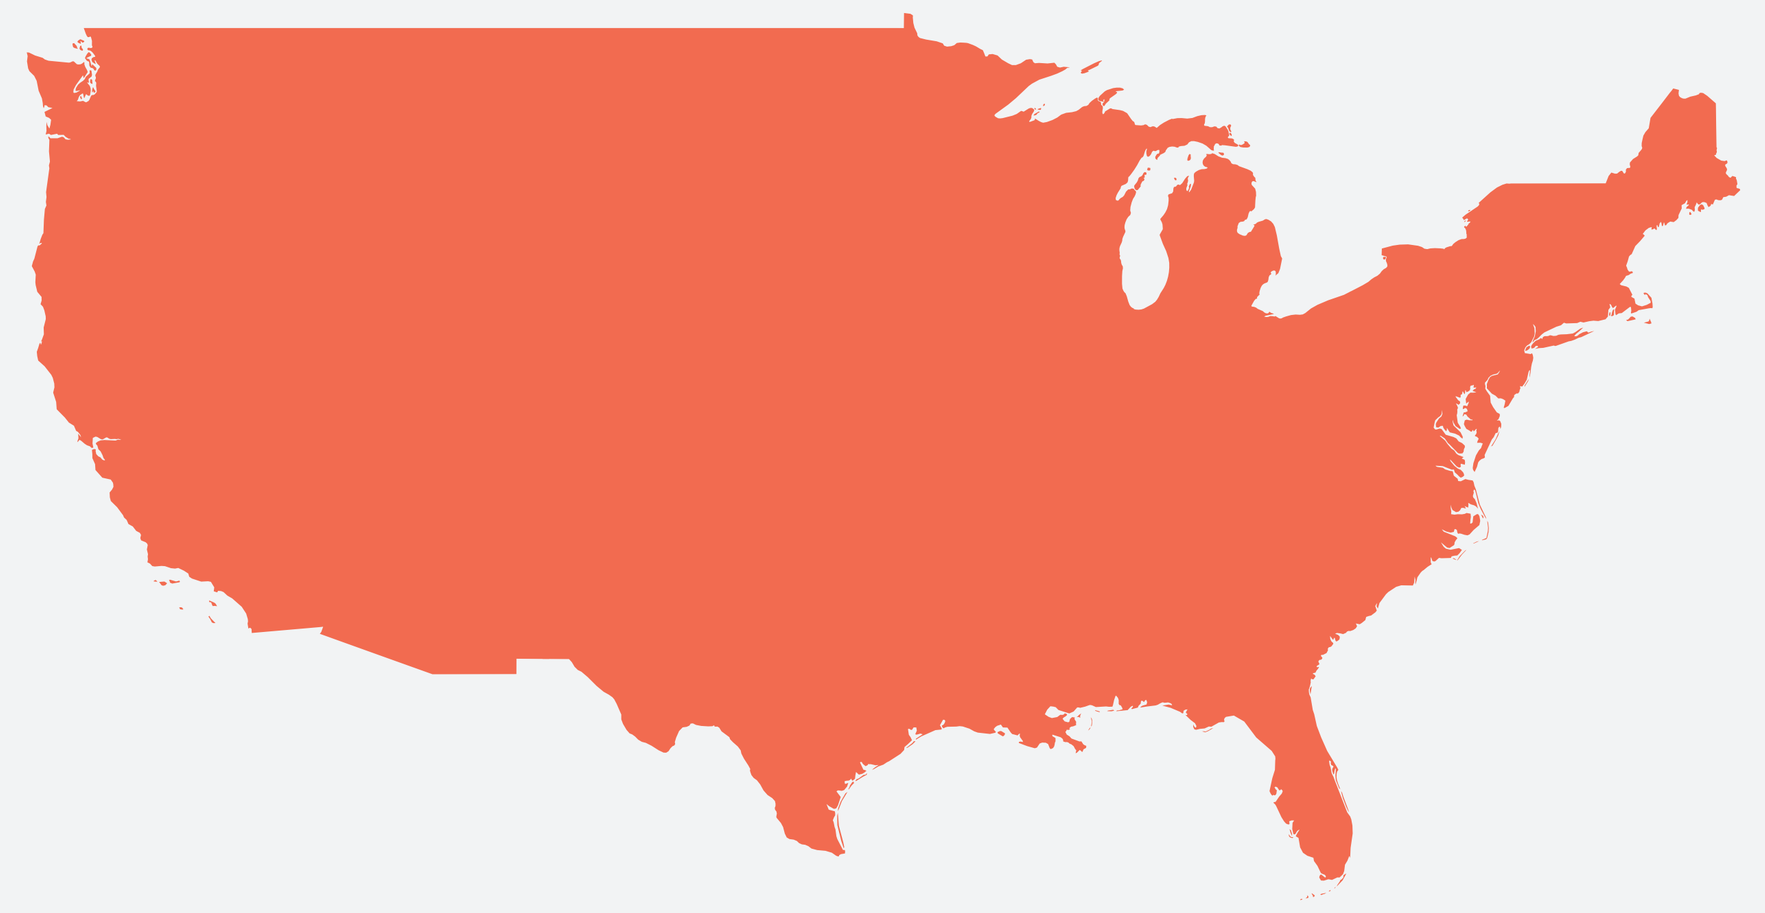

UNITED STATES

- Torrance, CA
- Aurora, CO
- Wilmington, DE
- Baltimore, MD
- Chevy Chase, MD
- Columbia, MO
- St. Louis, MO
- Durham, NC
- Nashville, TN
- Dallas, TX
- Madison, WI

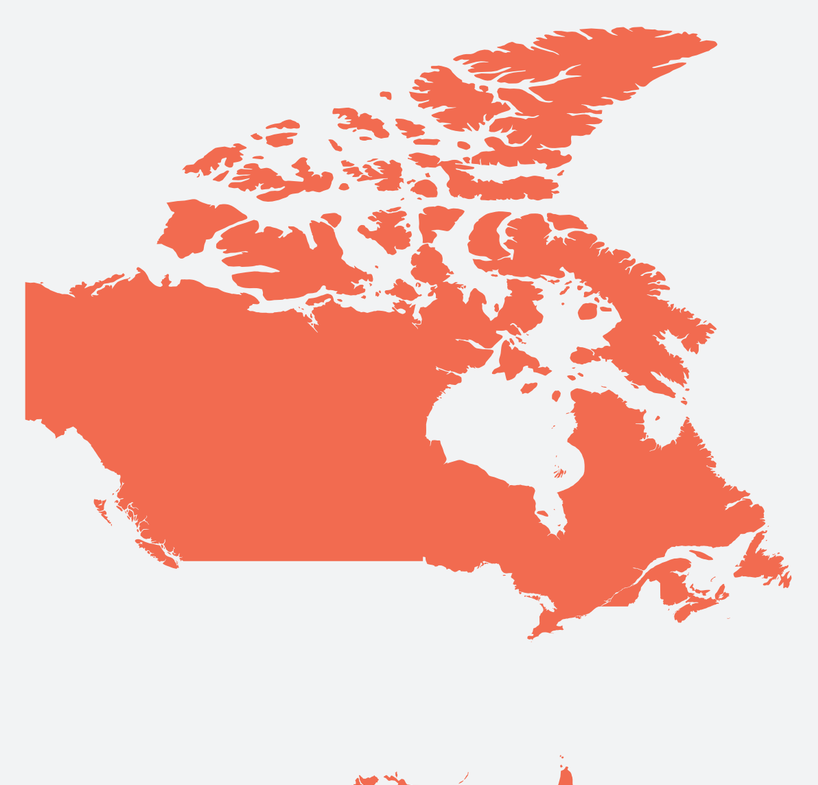

CANADA

- Toronto
- Edmonton
- Ottawa
- Montreal

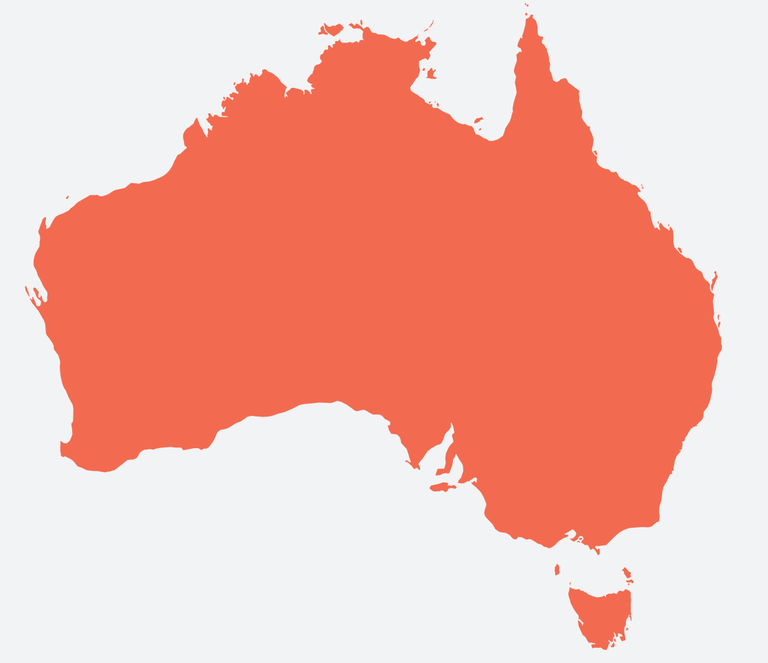

AUSTRALIA

- Parkville

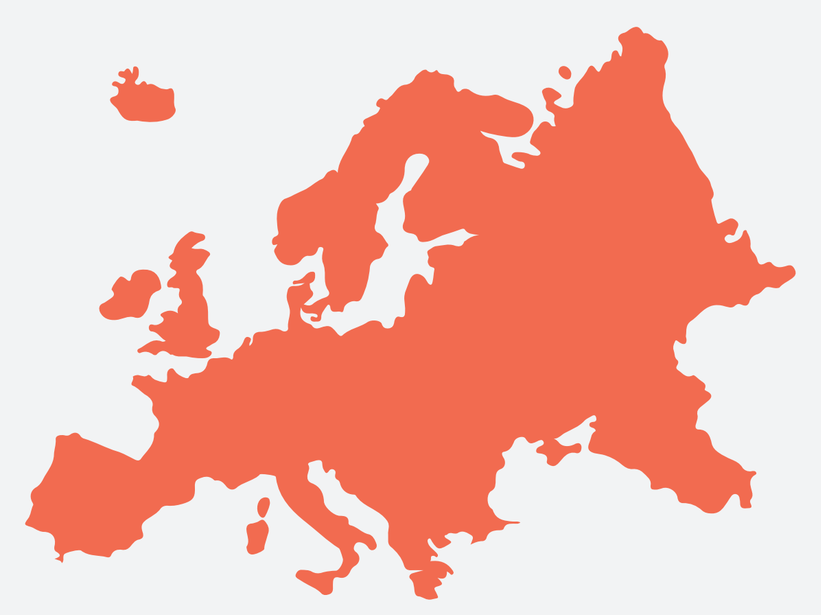

EUROPE

- Paris, France
- Utrecht, Netherlands
- Vitoria, Spain
- Madrid, Spain
- Stockholm, Sweden

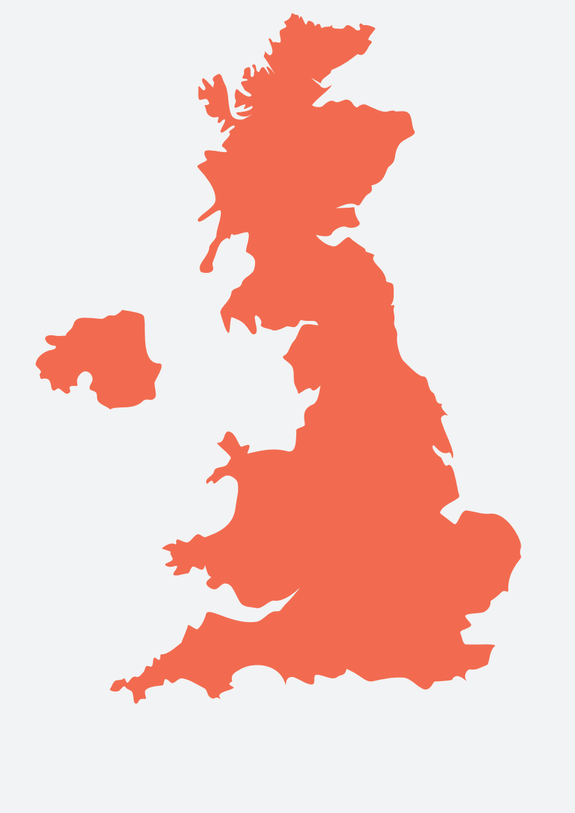

UK

- Sheffield
- London
 - Great Ormond Street Hospital
 - Guy's and St Thomas' NHS Foundation Trust

Site list reflects participating centers as of September 25, 2026.

Abbreviations

ACH, achondroplasia; AE, adverse event; AGV, annualized growth velocity; $AUC_{0\text{-last}}$, area under the curve from time zero to the last measurable concentration; AUC_{tau}, area under the curve from time zero to tau; $AUC_{0-\infty}$, area under the curve from time zero to infinity; CL/F, apparent clearance; C_{max}, maximum plasma concentration; CNP, C-type natriuretic peptide; FGFR, fibroblast growth factor receptor; Hx, history; mo, month; QD, once daily; $t_{½}$, half-life; T_{max}, time to maximum concentration; Vd/F, apparent volume of distribution.

References

1. Pauli RM. *Orphanet J Rare Dis.* 2019;14(1):1. 2. Ornitz DM and Legeai-Mallet L. *Dev Dyn.* 2017;246(4):291-309. 3. Chamarthi VS, et al. Achondroplasia. Treasure Island (FL): *StatPearls Publishing*; 2026. 4. Monsonego-Ornan E, et al. *Mol Cell Biol.* 2000;20(2):516-522. 5. Tyra Biosciences, Inc. Corporate presentation. December 2025. 6. Hoover-Fong J, et al. *Pediatrics.* 2020;145(6):e20201010. 7. Hecht JT, et al. *Am J Med Genet.* 1989;32(4):528-535. 8. Savarirayan R, et al. *Lancet.* 2020;396(10252):684-692. 9. Savarirayan R, et al. *JAMA Pediatr.* 2026;180(1):18-25. 10. Savarirayan R, et al. *N Engl J Med.* 2025;392(9):865-874. *11.* Starrett JH, et al. *Mol Cancer Ther.* 2026;25(3):408-415. 12. Hudkins RL, et al. *J Med Chem.* 2024;67(18):16737-16756. 13. National Library of Medicine. NCT06842355. Accessed September 25, 2026. https://clinicaltrials.gov/study/NCT06842355.

Acknowledgments

The authors would like to thank the patients, caregivers, and the BEACH301 investigators and study personnel at study sites. Medical writing assistance was provided by Federica Rinaldi, PhD, of MEDiSTRAVA (Yardley).
This study is funded by Tyra Biosciences.

Sponsor Contact Information

TyraClinicalTrials@tyra.bio | Tyra Biosciences, Inc. | Carlsbad, CA, USA

Presented at the American Society for Bone and Mineral Research (ASBMR) Annual Meeting; October 9–12, 2026; Boston, MA, USA

Content of this poster is copyrighted by Tyra Biosciences, Inc. Permission is required for reuse.